Gait analysis in a porcine model of chronic Parkinson disease established by continuous MPTP intoxication

P Mogensen[1], MS Nielsen[2], AN Glud[2], A Møller[3], D Bender[4], D Doudet[3], JC Sørensen[5], CR Bjarkam[2,5]

[1]Hammel Neurocenter, Voldbyvej 15, 8450 Hammel, Denmark; [2]Institute of Anatomy, Faculty of Health Sciences, University of Aarhus, Wilh. Meyers Allé bygn. 3, 8000 Århus C, Denmark; [3]CFIN, Aarhus University Hospital, Nørrebrogade 44, 8000 Århus C, Denmark; [4]PET Center, Aarhus University Hospital, Nørrebrogade 44, 8000 Århus C, Denmark; [5]Department of Neurosurgery, Aarhus University Hospital, Nørrebrogade 44, 8000 Århus C, Denmark

Introduction

Parkinson disease (PD) is a serious neurological disorder resulting from an excessive loss of dopaminergic neurons in the substantia nigra (SN). Animal models have been very helpful in gaining a better understanding of disease mechanisms and in the developing of new treatments for the disease. However, most animal models are based on acute neuron loss and thus do not mimic the progressive neuron loss seen in patients. This may be an important reason why preclinical studies of neuroprotection have so far not been predictive of results in patients. In addition, high costs and ethical concerns are making it increasable difficult to perform experiments on primates. To address these obstacles, we wanted to develop a functional non-primate large animal model of chronic PD based on continuous 1-methyl-4-phenyl-1,2,3,6-tetrahydropyridine (MPTP) intoxication in the Göttingen minipig.

Materials and methods

Eighteen Göttingen minipigs (figure 1) were used for the study. Fourteen Göttingen minipigs were implanted subcutaneously with infusion pumps for continuous intramuscular MPTP delivery at daily doses of 2-24 mg's pr. day. Four animals served as normal controls. The animals were followed for 11 weeks, during which they were observed and scored behaviorally on motility, coordination, rigidity, chewing, and vocalization.

All animals underwent digital movement analysis preoperatively, 4 and 11 weeks postoperatively using an infrared 3-D computerized Vicon system to measure the temporospatiale parameters of gait. Gait velocity, step length, gait cycle, raise of the leg from the floor, single stand and double support phase (hind legs) were measured (figure 2).

At the time of sacrifice, fresh striatal biopsies were examined for dopamine content using HPLC.

The animals were transcardially perfused with 4% PFA, the brain removed, paraffin embedded, cut 30 μm thick and examined histopathologically and stereologically for total number of tyrosine hydroxylase(TH)-positive neurons in the SN.

Results

All MPTP-intoxicated animals developed symptoms of PD after 10-14 days. We observed decreased motility (bradykinesia/akinesia), coordination difficulties starting in the hind limbs, chewing difficulties and in some cases abnormal vocalization. However, the symptoms were mild and transient in the 2-12 mg groups. The 18 mg group developed moderate and stable symptoms, whereas the animals in the 24 mg group were severely parkinsonian after just 11 days of intoxication and had their pumps turned off.

Four weeks postoperatively the gait velocity in the 12 mg group was normal. At that time the 18 mg group had a significant decrease in their gait velocity. After 11 weeks of intoxication both groups showed a decreased gait velocity (figure 3). Gait cycle showed the same pattern. After 4 weeks of intoxication, gait cycle was decreased in the 18 mg animals (figure 2). After 11 weeks both groups walked with a decreased gait cycle. There was a small, but significant decrease in step length after 11 weeks in the 18 mg group (figure 4). We know from our studies of normal pigs, that there is a linier relationship between step length and gait velocity.

Thus there were no changes in step length. The pigs raised the limb from the floor in a normal manner. However, again we know from studies of normal pigs, that there is a linier relationship between raise of limb and gait velocity. We therefore found a clear dose dependent increase in raise of limb as a function of daily MPTP dose.

After sacrifice, HPLC showed significantly decreased levels of dopamine in the 12, 18 and 24 mg animals. Neuropathologically, neuron loss in the SN was evident in the 12-24 mg animels. This finding was confirmed by stereological estimation of the total number of TH-positive neurons in the SN, revealing a dose-response relation between the number of neurons left and the dose of MPTP.

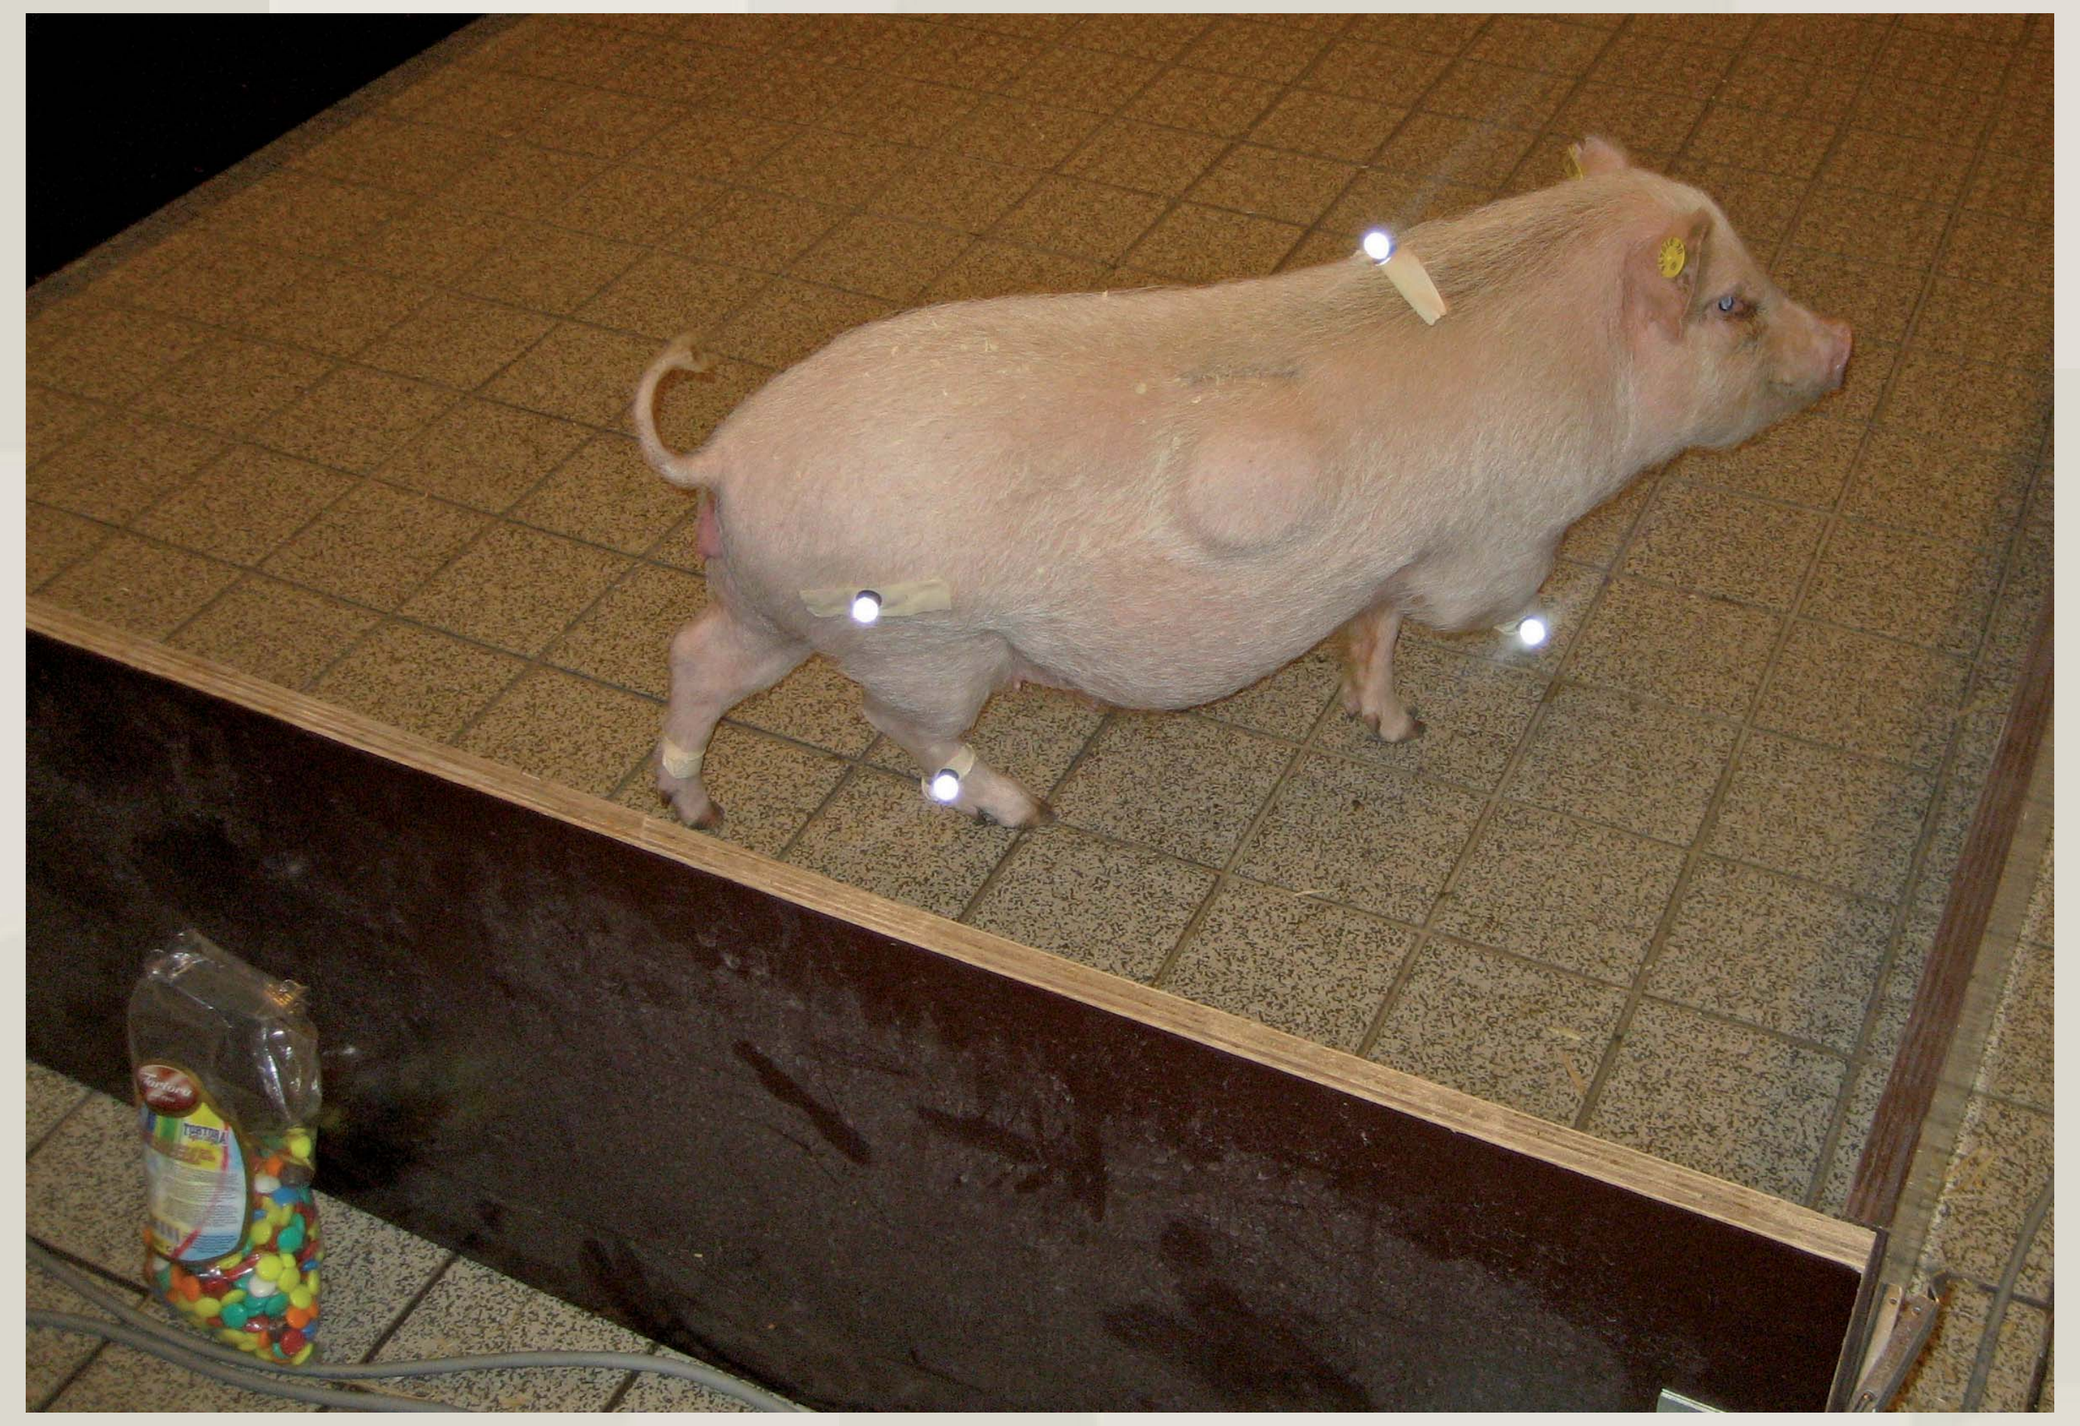

Figure 1:
The digital gait analysis. Six infrared Vicon cameras were placed around a fenced area. The pigs were trained to walk back and forth, being offered chocolate treats. With this procedure, we obtained objective measurements of gait velocity.

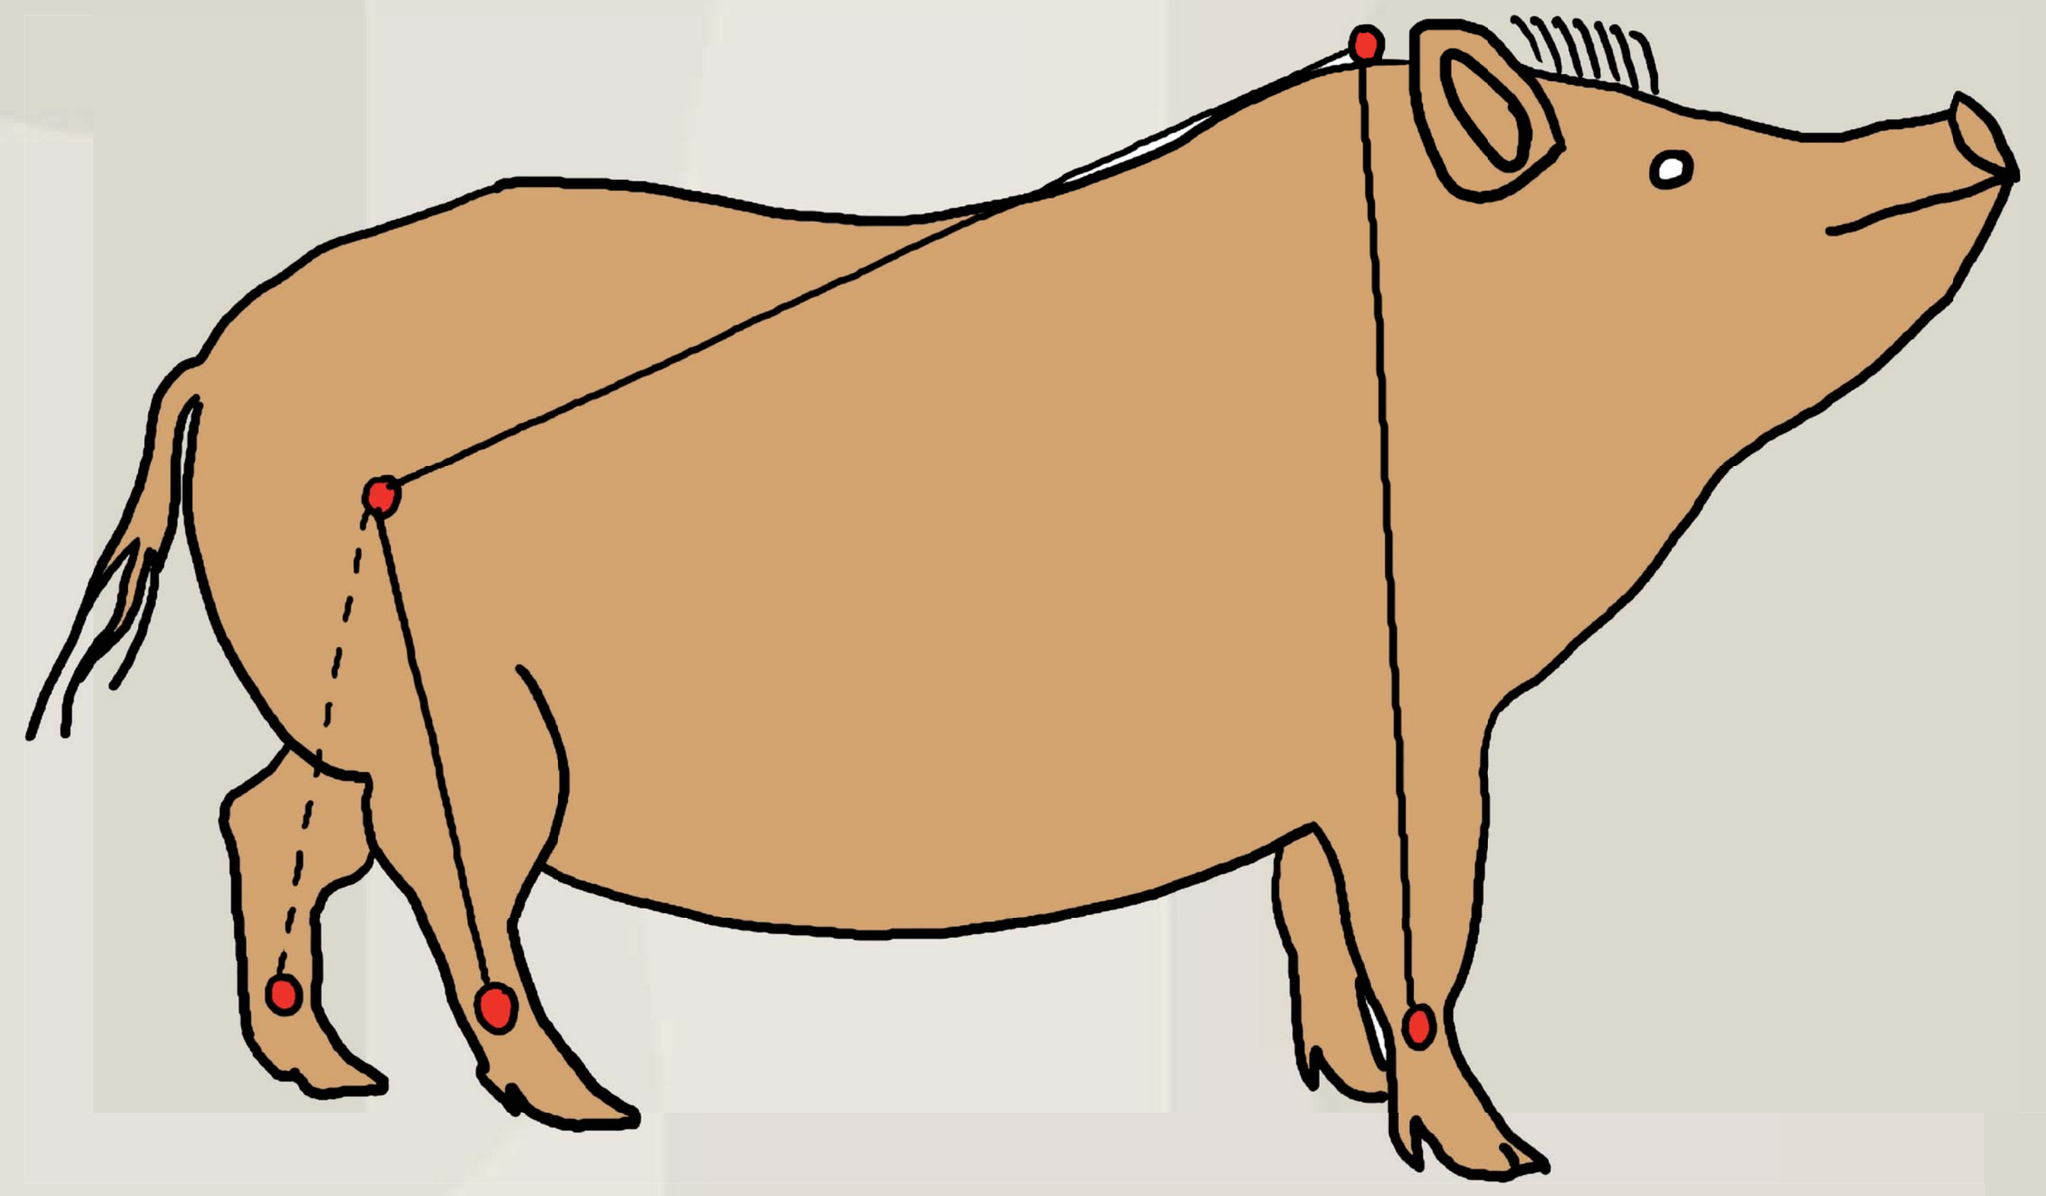

Figure 2:
Drawings of the pig with the 5 reflecting markers, as they are seen in the computer.

Acknowledgements

We greatly acknowledge the financial support from The Danish Medical Research Council, The Danish Medical Association/ Sven Aage Wacherhausen Nielsen Foundation, The Lundbeck Foundation and the Novo Nordisk Foundation.

Figure 3

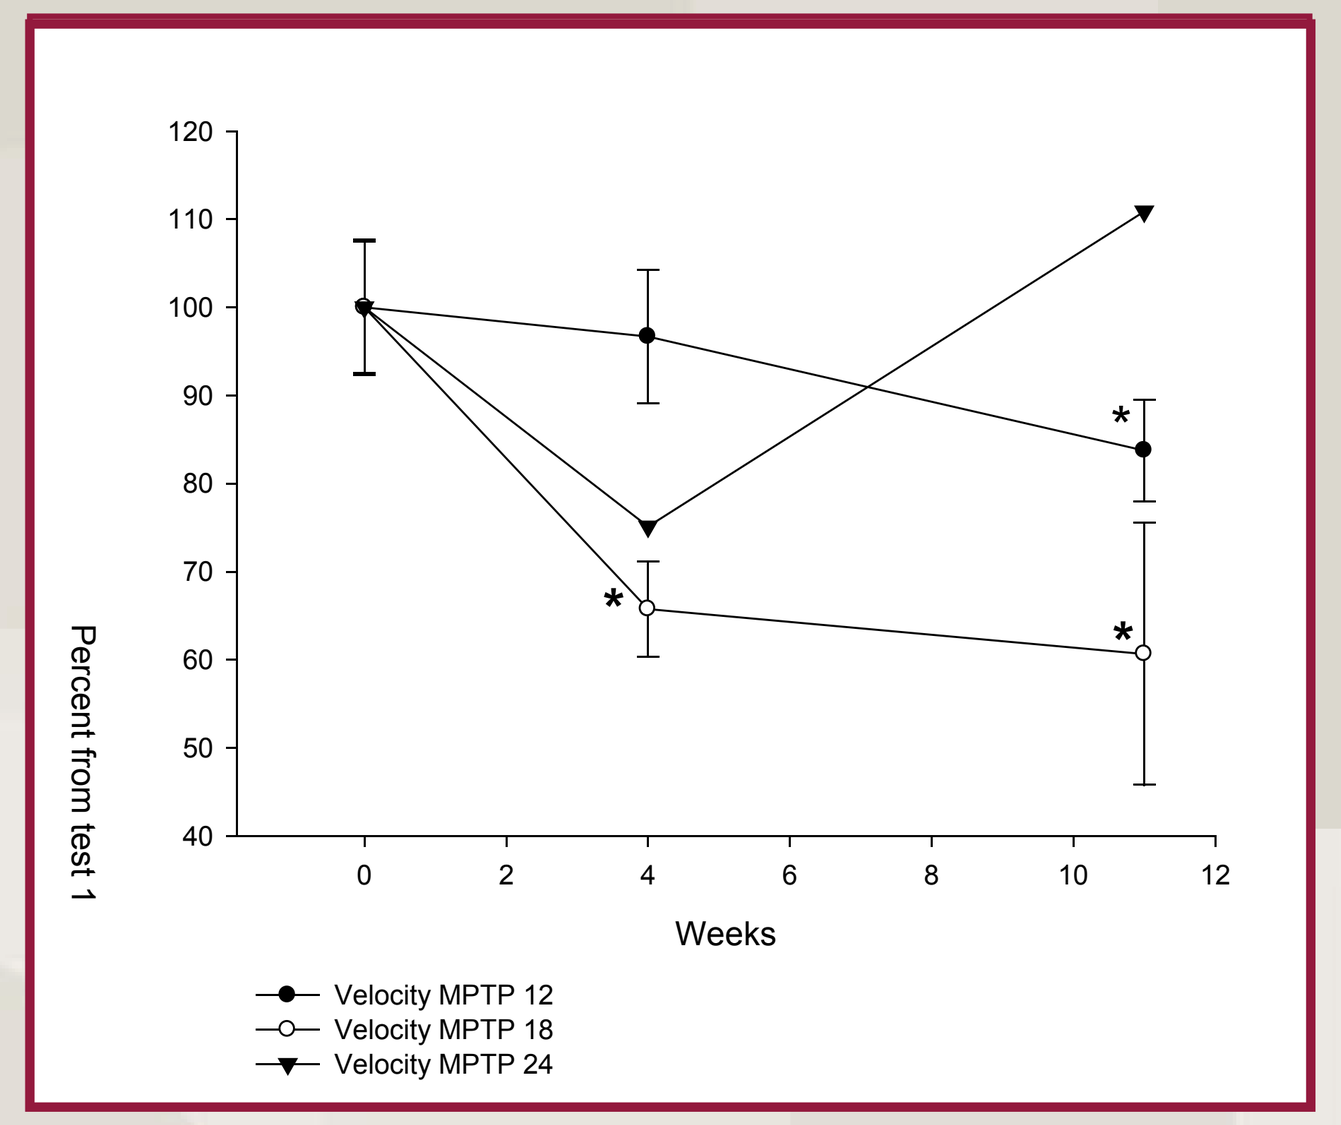
Percent from test 1
Weeks
Velocity MPTP 12
Velocity MPTP 18
Velocity MPTP 24

Gait velocity as a function of time.
The gait velocity decrease as a function of given MPTP/day over time. Twelve mg of MPTP induced a decrease in gait velocity after 11 weeks. After only 4 weeks there was a decrease in the 18 mg group. The 24 mg animal had a decreased gait cycle 4 week, but not 12 weeks after operation. It had the pump shut of, after only 11 days because of symptoms of acute intoxication.

Figure 4

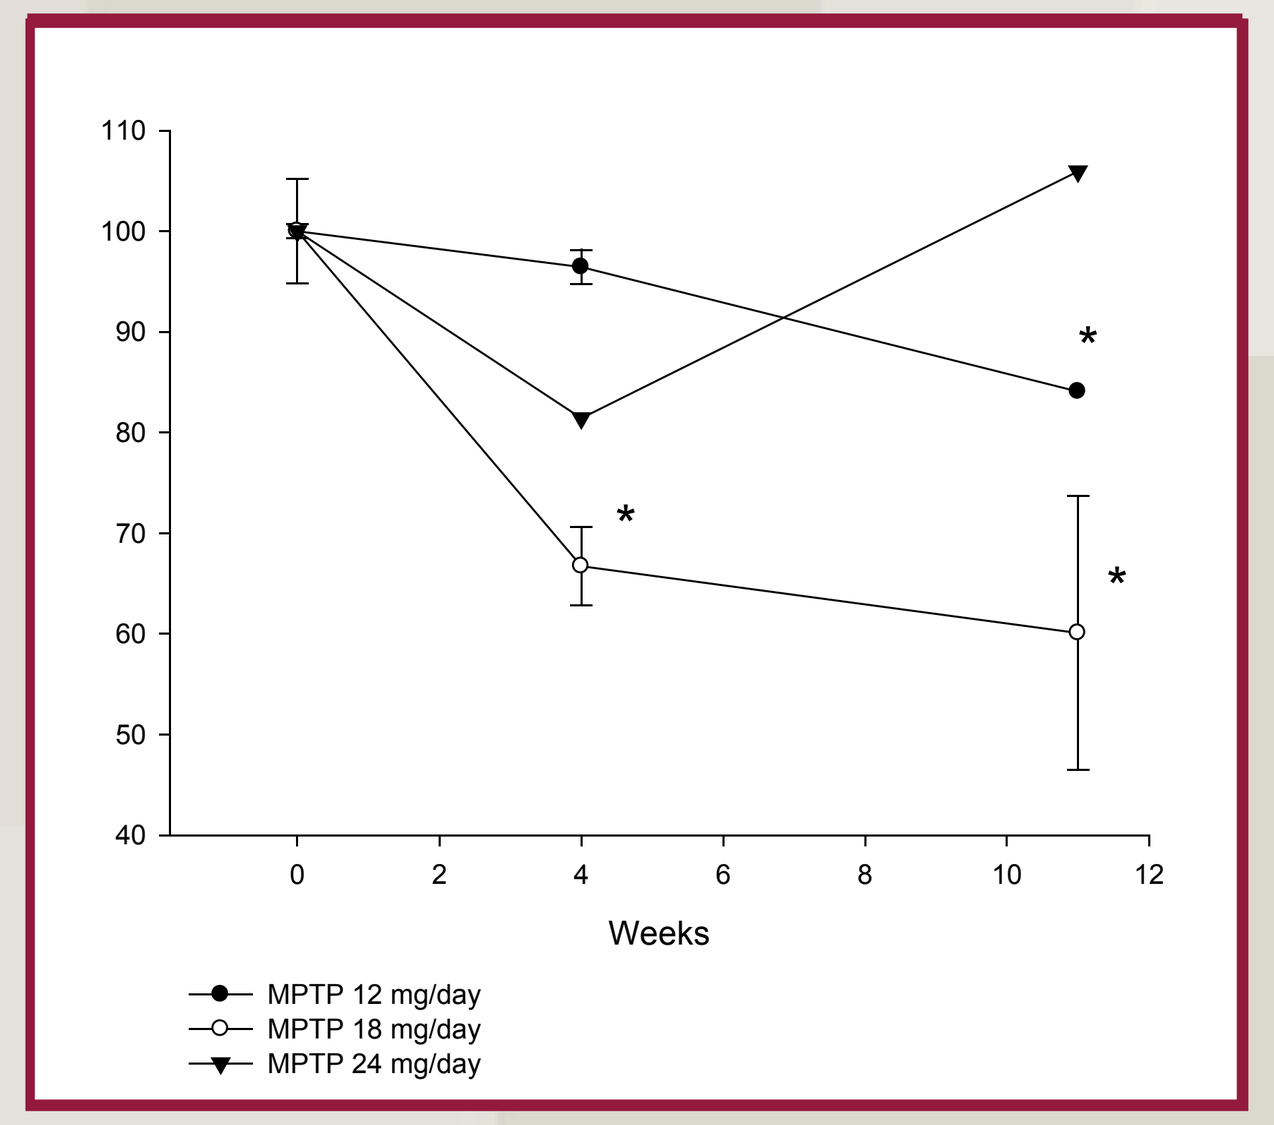
Weeks
MPTP 12 mg/day
MPTP 18 mg/day
MPTP 24 mg/day

Gait cycle of the right hind leg as a function of time.
After 4 weeks there was a decrease in the 18 mg group. In the 12 mg group the decrease was first seen after 11 weeks. Again the 24 mg/day animal shows signs of acute intoxication.

Figure 5:

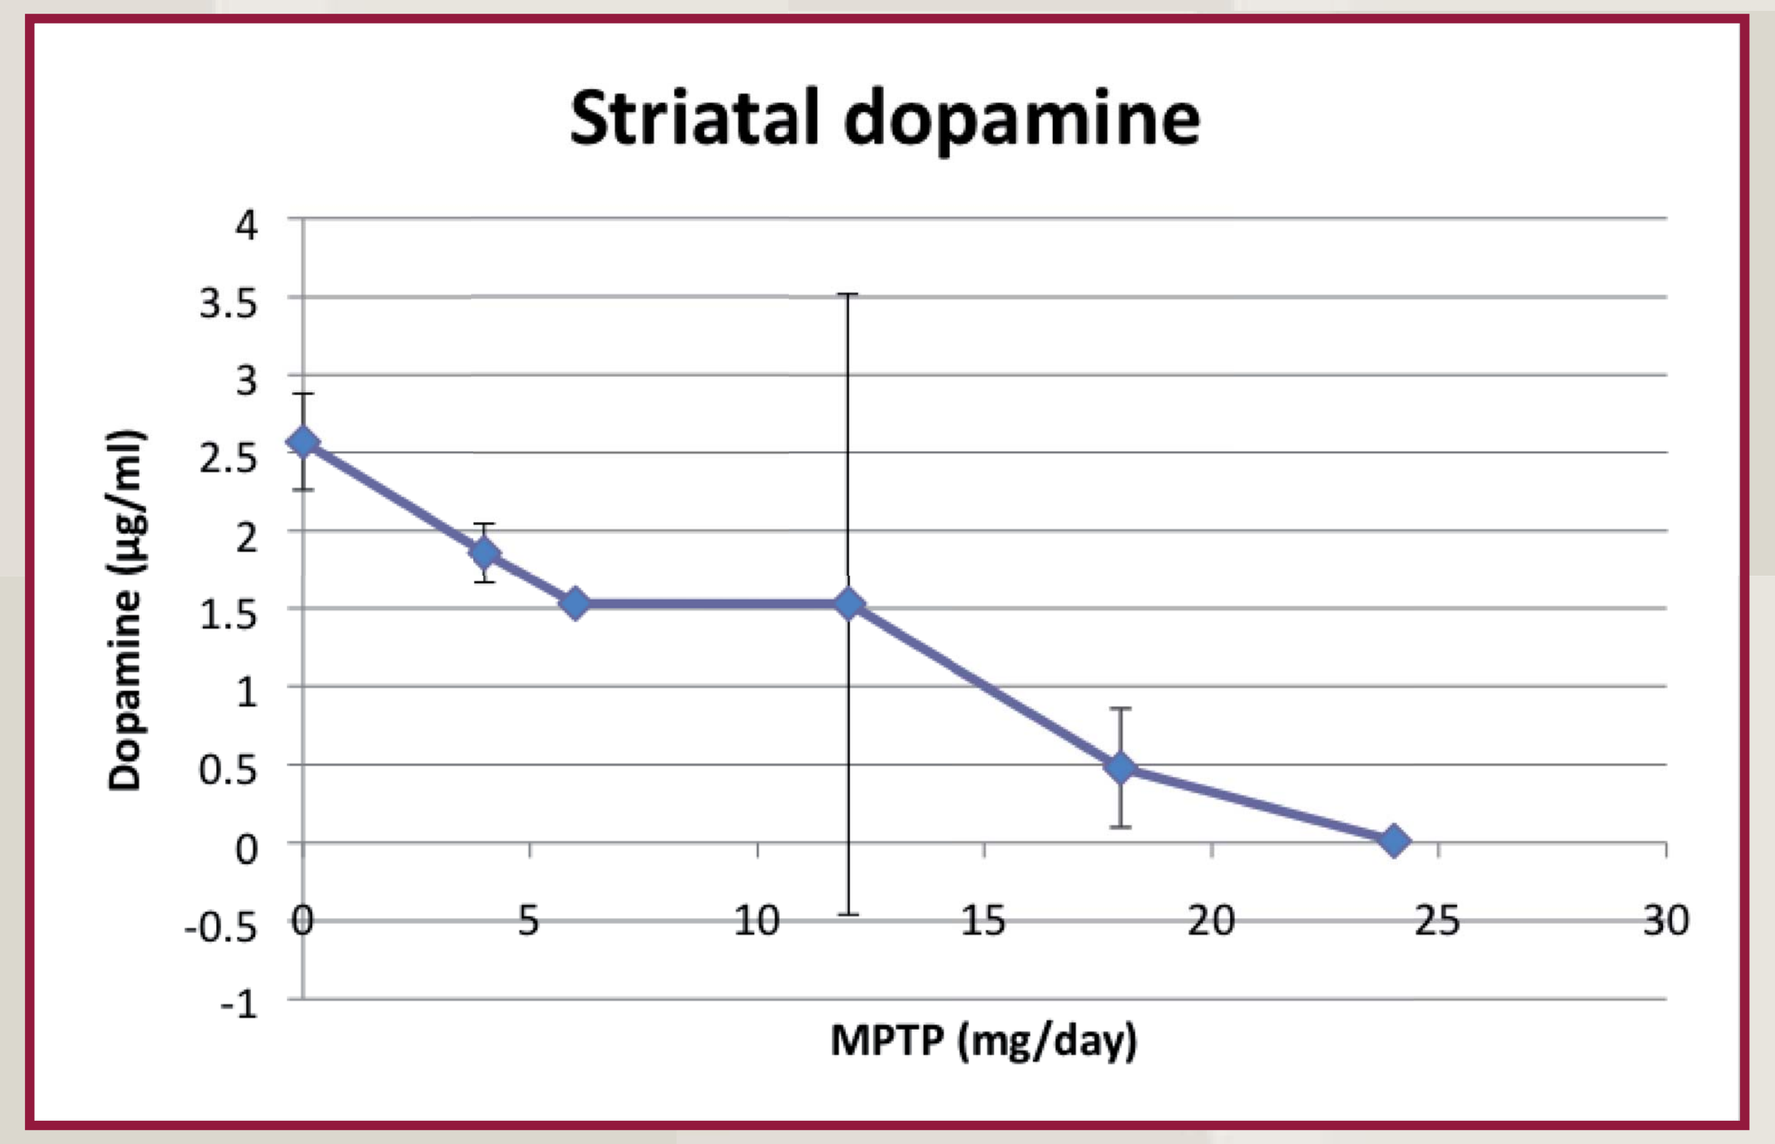
Striatal dopamine
Dopamine (µg/ml)
MPTP (mg/day)

HPLC of dopamine content in the striatum. Significantly decreased levels were seen in 18 and 24 mg animals. The large standard deviations are due to small sample sizes (few animals included) and the low values.

Figure 6:

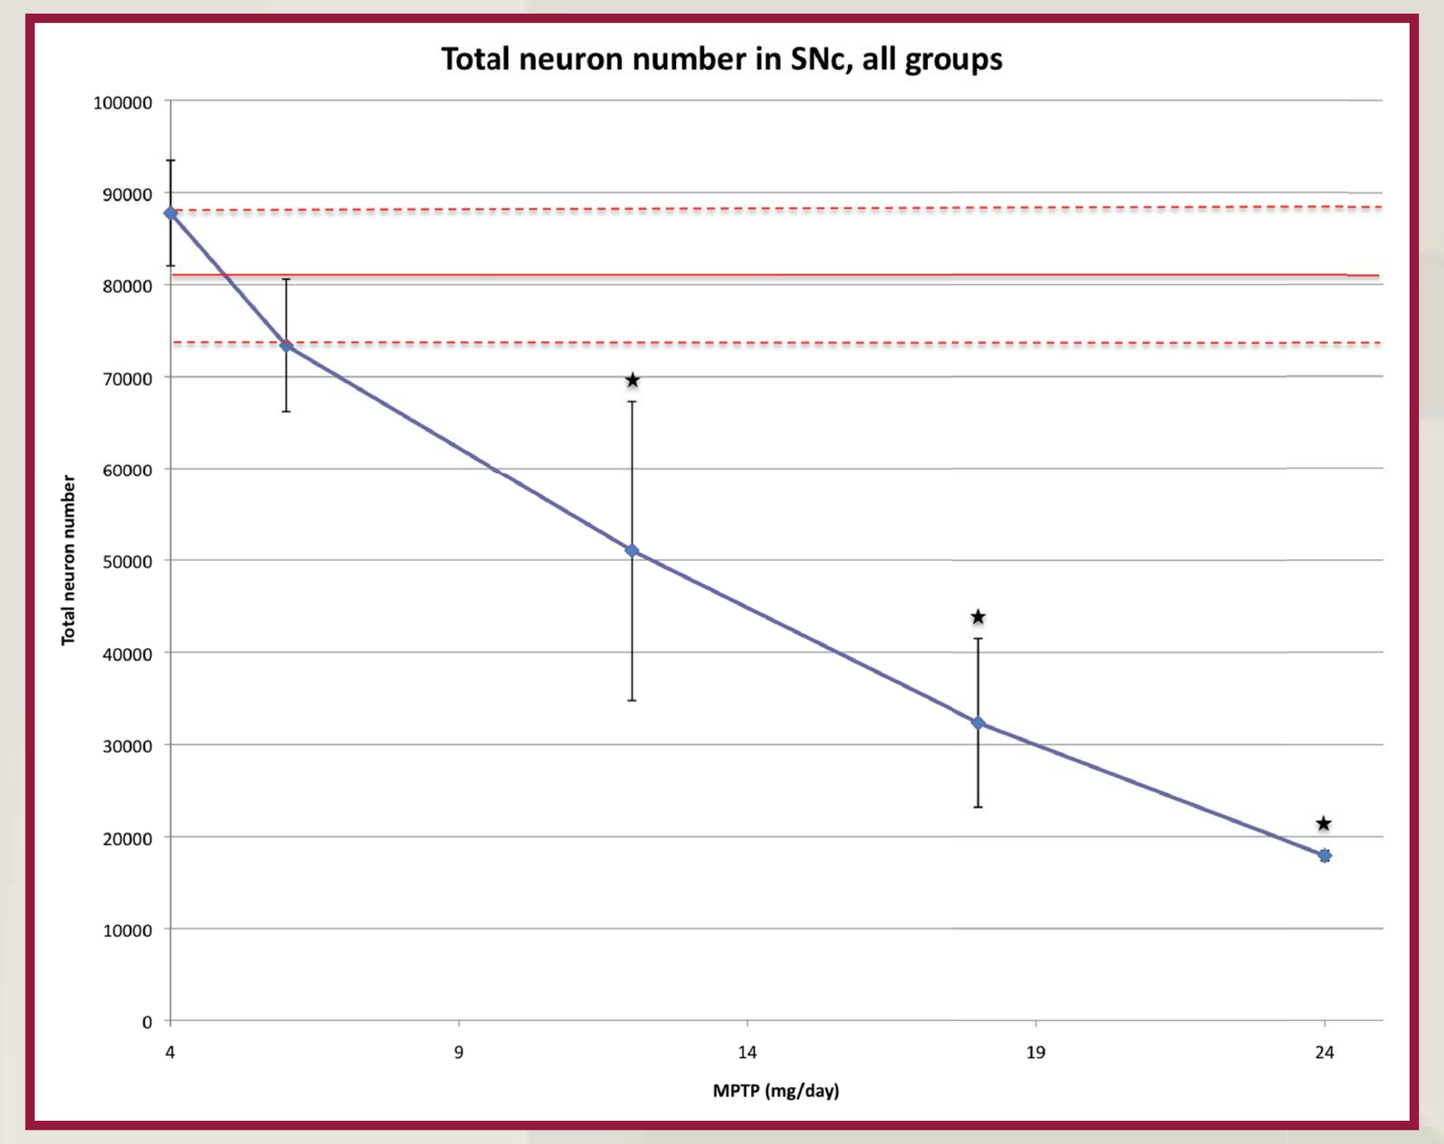
Total neuron number in SNc, all groups
Total neuron number
MPTP (mg/day)

Stereological analysis of the total neuron number in the SN employing the optical dissector method in TH-stained sectors. A statistically significant neuron loss was seen in the 12, 18 and 18 mg groups. Again, a clear dose-response relation was evident.

Conclusion

We conclude that it is possible to perform chronic MPTP-intoxication in Göttingen minipigs and thereby obtain a robust functional gait model of Parkinson disease.

This study showed that with continuous infusion of MPTP it is possible to develop PD symptoms in the Göttingen minipig ranging from mild and transient to moderate and stable and finally severe, mimicking the acute intoxication. We found a clear dose response relation between daily MPTP dose and symptoms, correlating with the gait analysis results.

MPTP-intoxicated Göttingen minipigs walked with decreased velocity. We show that this decreased is caused by a decreased in gait cycle and not by change in step length.

We found a clear dose response relation between daily MPTP dose and dopamine content in the striatum (figure 5) and stereological cell count in the SN (figure 6). We found 18 mg MPTP/day to be the optimal dose as these animals presented with stable symptoms.

The Göttingen minipig thus offers an exciting alternative to primates as a large animal model and may prove advantageous future in neuroprotective studies.